Pharmacokinetics of alkylamides and caffeic acid conjugates from Echinacea

A. Matthias[1], K. G. Penman[1], R.S. Addison[3], R.G. Dickinson[3], K.M. Bone[1,2], & R.P. Lehmann[1]

[1]MediHerb Research Laboratories, University of Queensland, Brisbane, 4072; [2]School of Health, University of New England, NSW 2351;
[3]Centre for Studies in Drug Disposition, University of Queensland, Royal Brisbane Hospital, Brisbane, 4029 Australia

INTRODUCTION

Relatively little is known about the bioavailability and pharmacokinetics of the alkylamides and caffeic acid conjugates found in echinacea. The major tetraene (2E,4E,8Z,10Z)-N-isobutyldodeca-2,4,8,10-tetraenamide (m/z = 247) has been shown to be present in plasma 1 hour after ingestion of an echinacea ethanolic liquid [1] but nothing is known about the bioavailability of the caffeic acid conjugates. A recent study using Caco-2 monolayers [2] has shown that Echinacea alkylamides should easily cross the intestinal barrier and be present in plasma while caffeic acid conjugates are unlikely to pass the intestinal barrier and thus not appear in plasma.
In this study, we have investigated the bioavailability and pharmacokinetics of alkylamides and caffeic acid conjugates after ingestion of Echinacea tablets (MediHerb Echinacea Premium Tablet). Bioavailability is the fraction of a given dose of compound which reaches the systemic circulation as intact compound. Pharmacokinetics is the time course of compound concentration in the body and has four phases - absorption, distribution, metabolism and excretion.

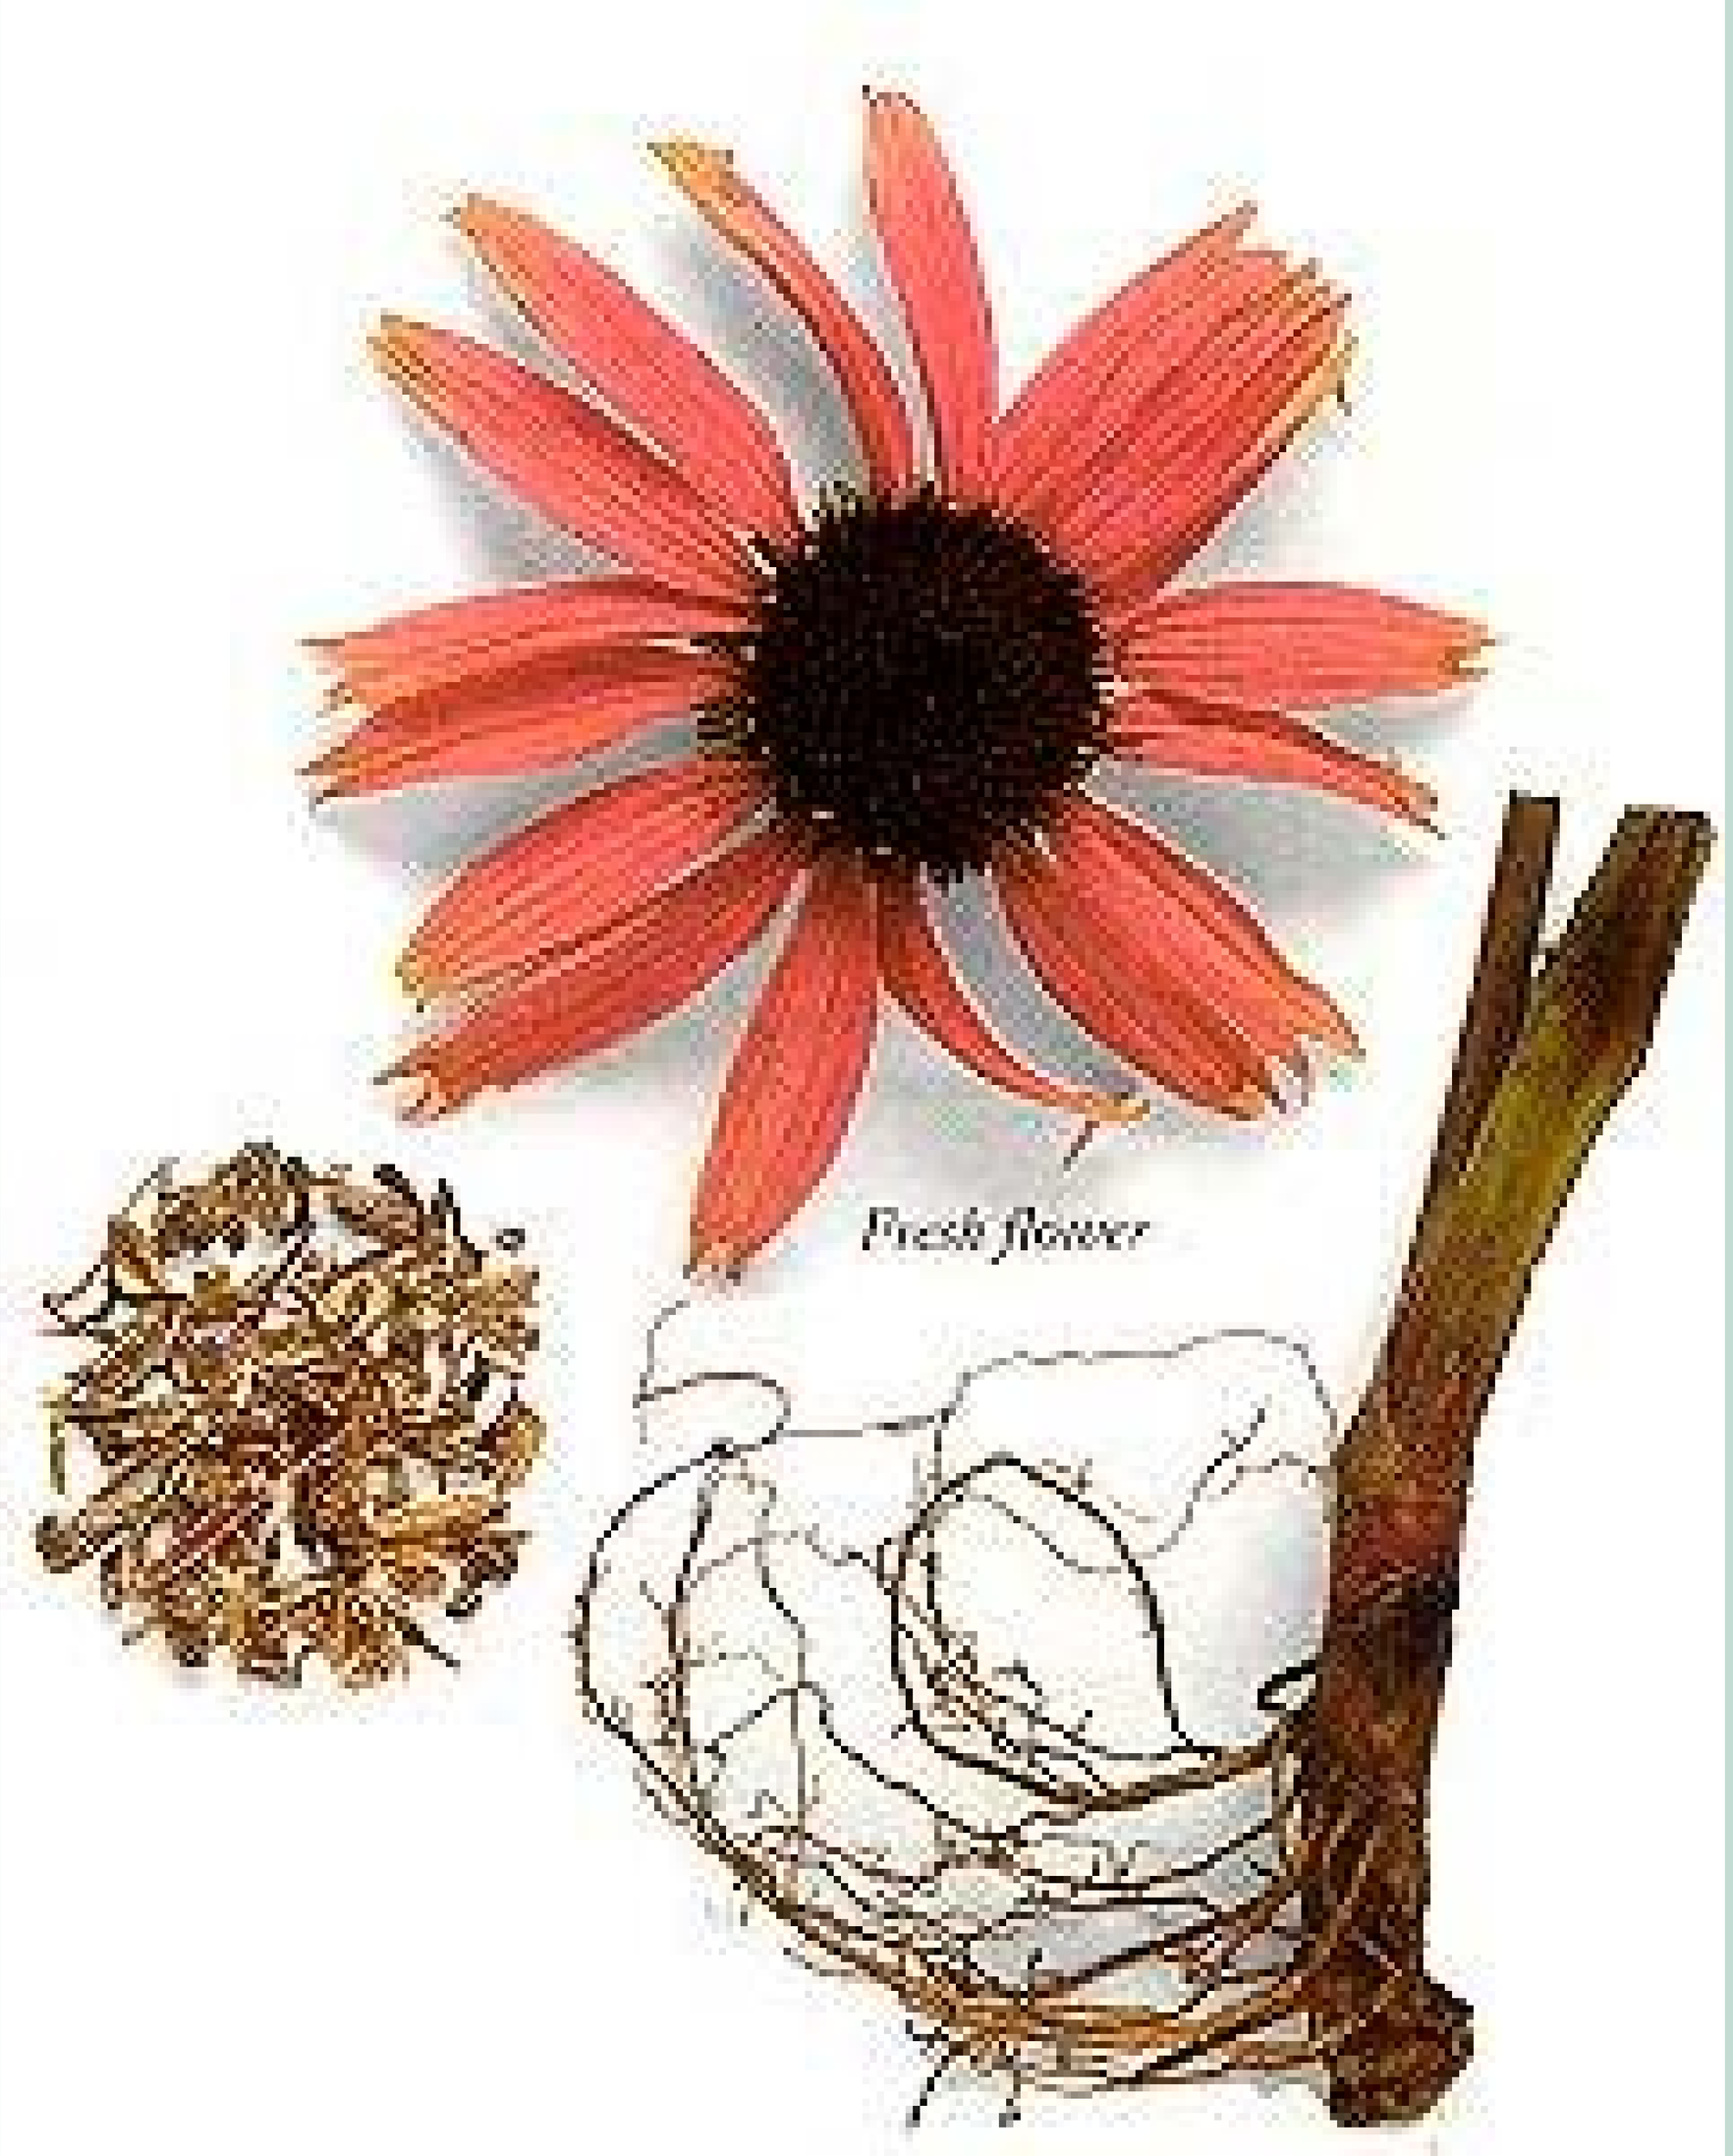
Fresh flower

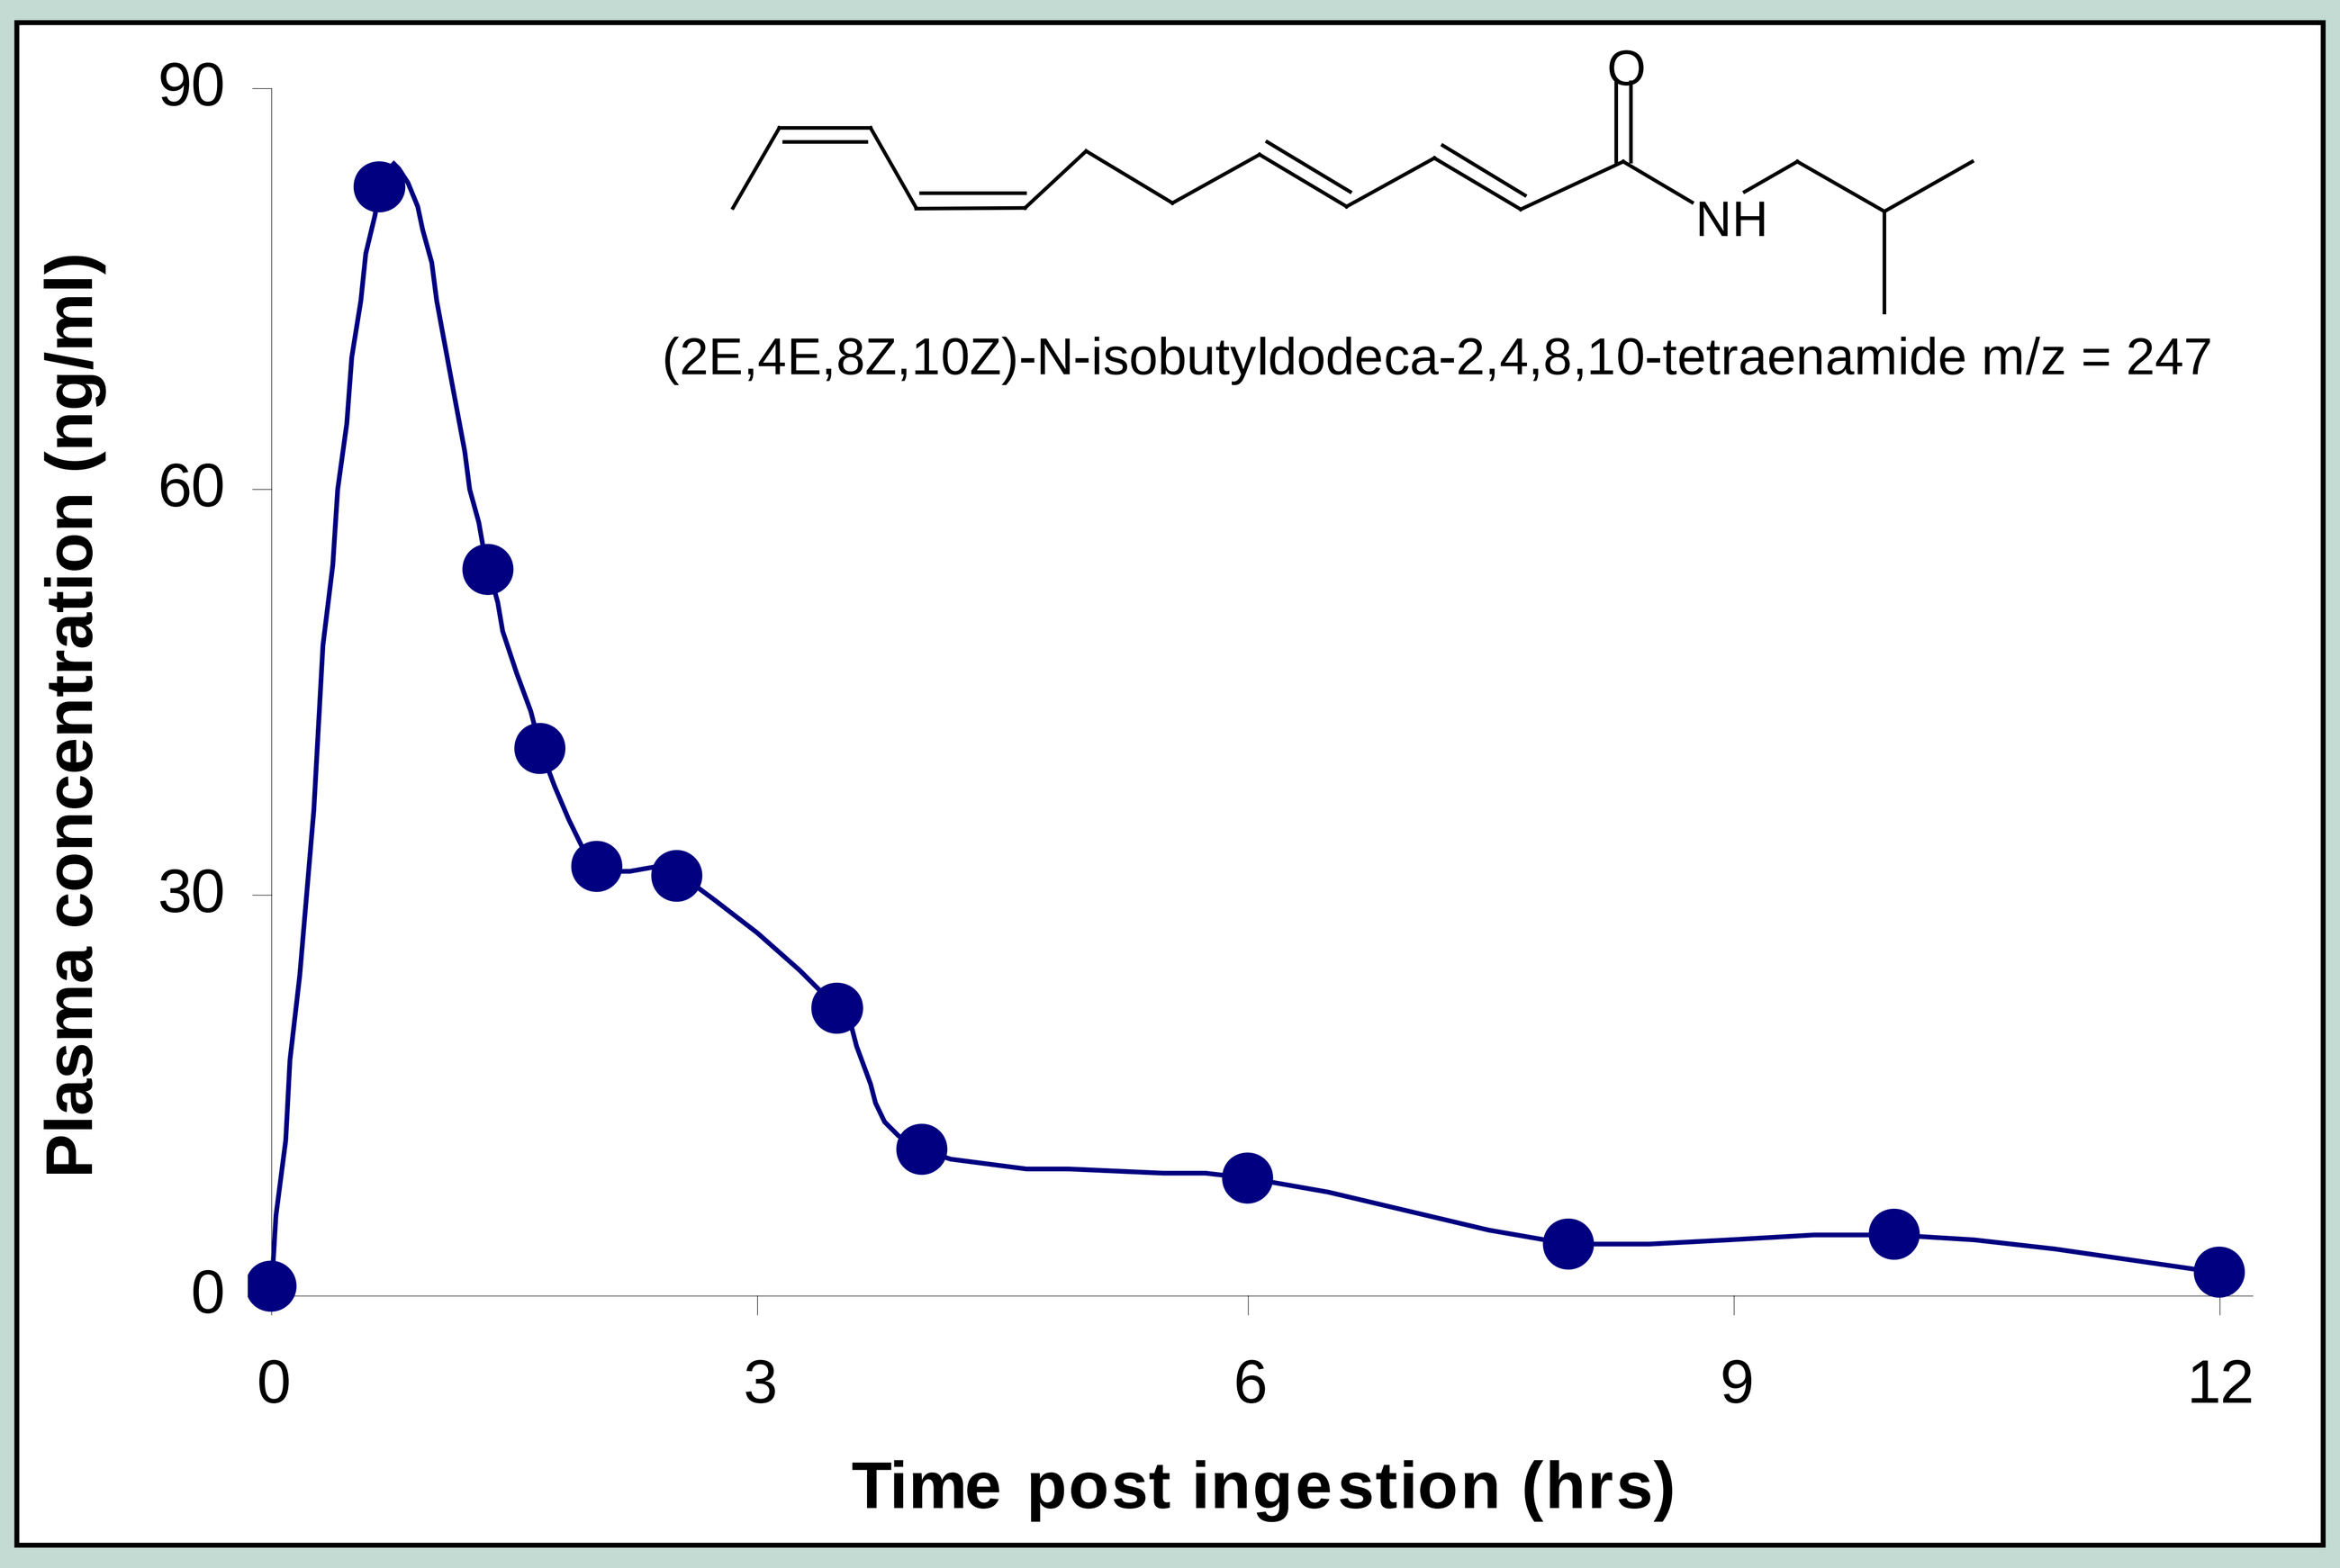
(2E,4E,8Z,10Z)-N-isobutyldodeca-2,4,8,10-tetraenamide m/z = 247
Plasma concentration (ng/ml)
0
30
60
90
0
3
6
9
12
Time post ingestion (hrs)

Figure 1: Plasma alkylamide (m/z = 247) concentration.

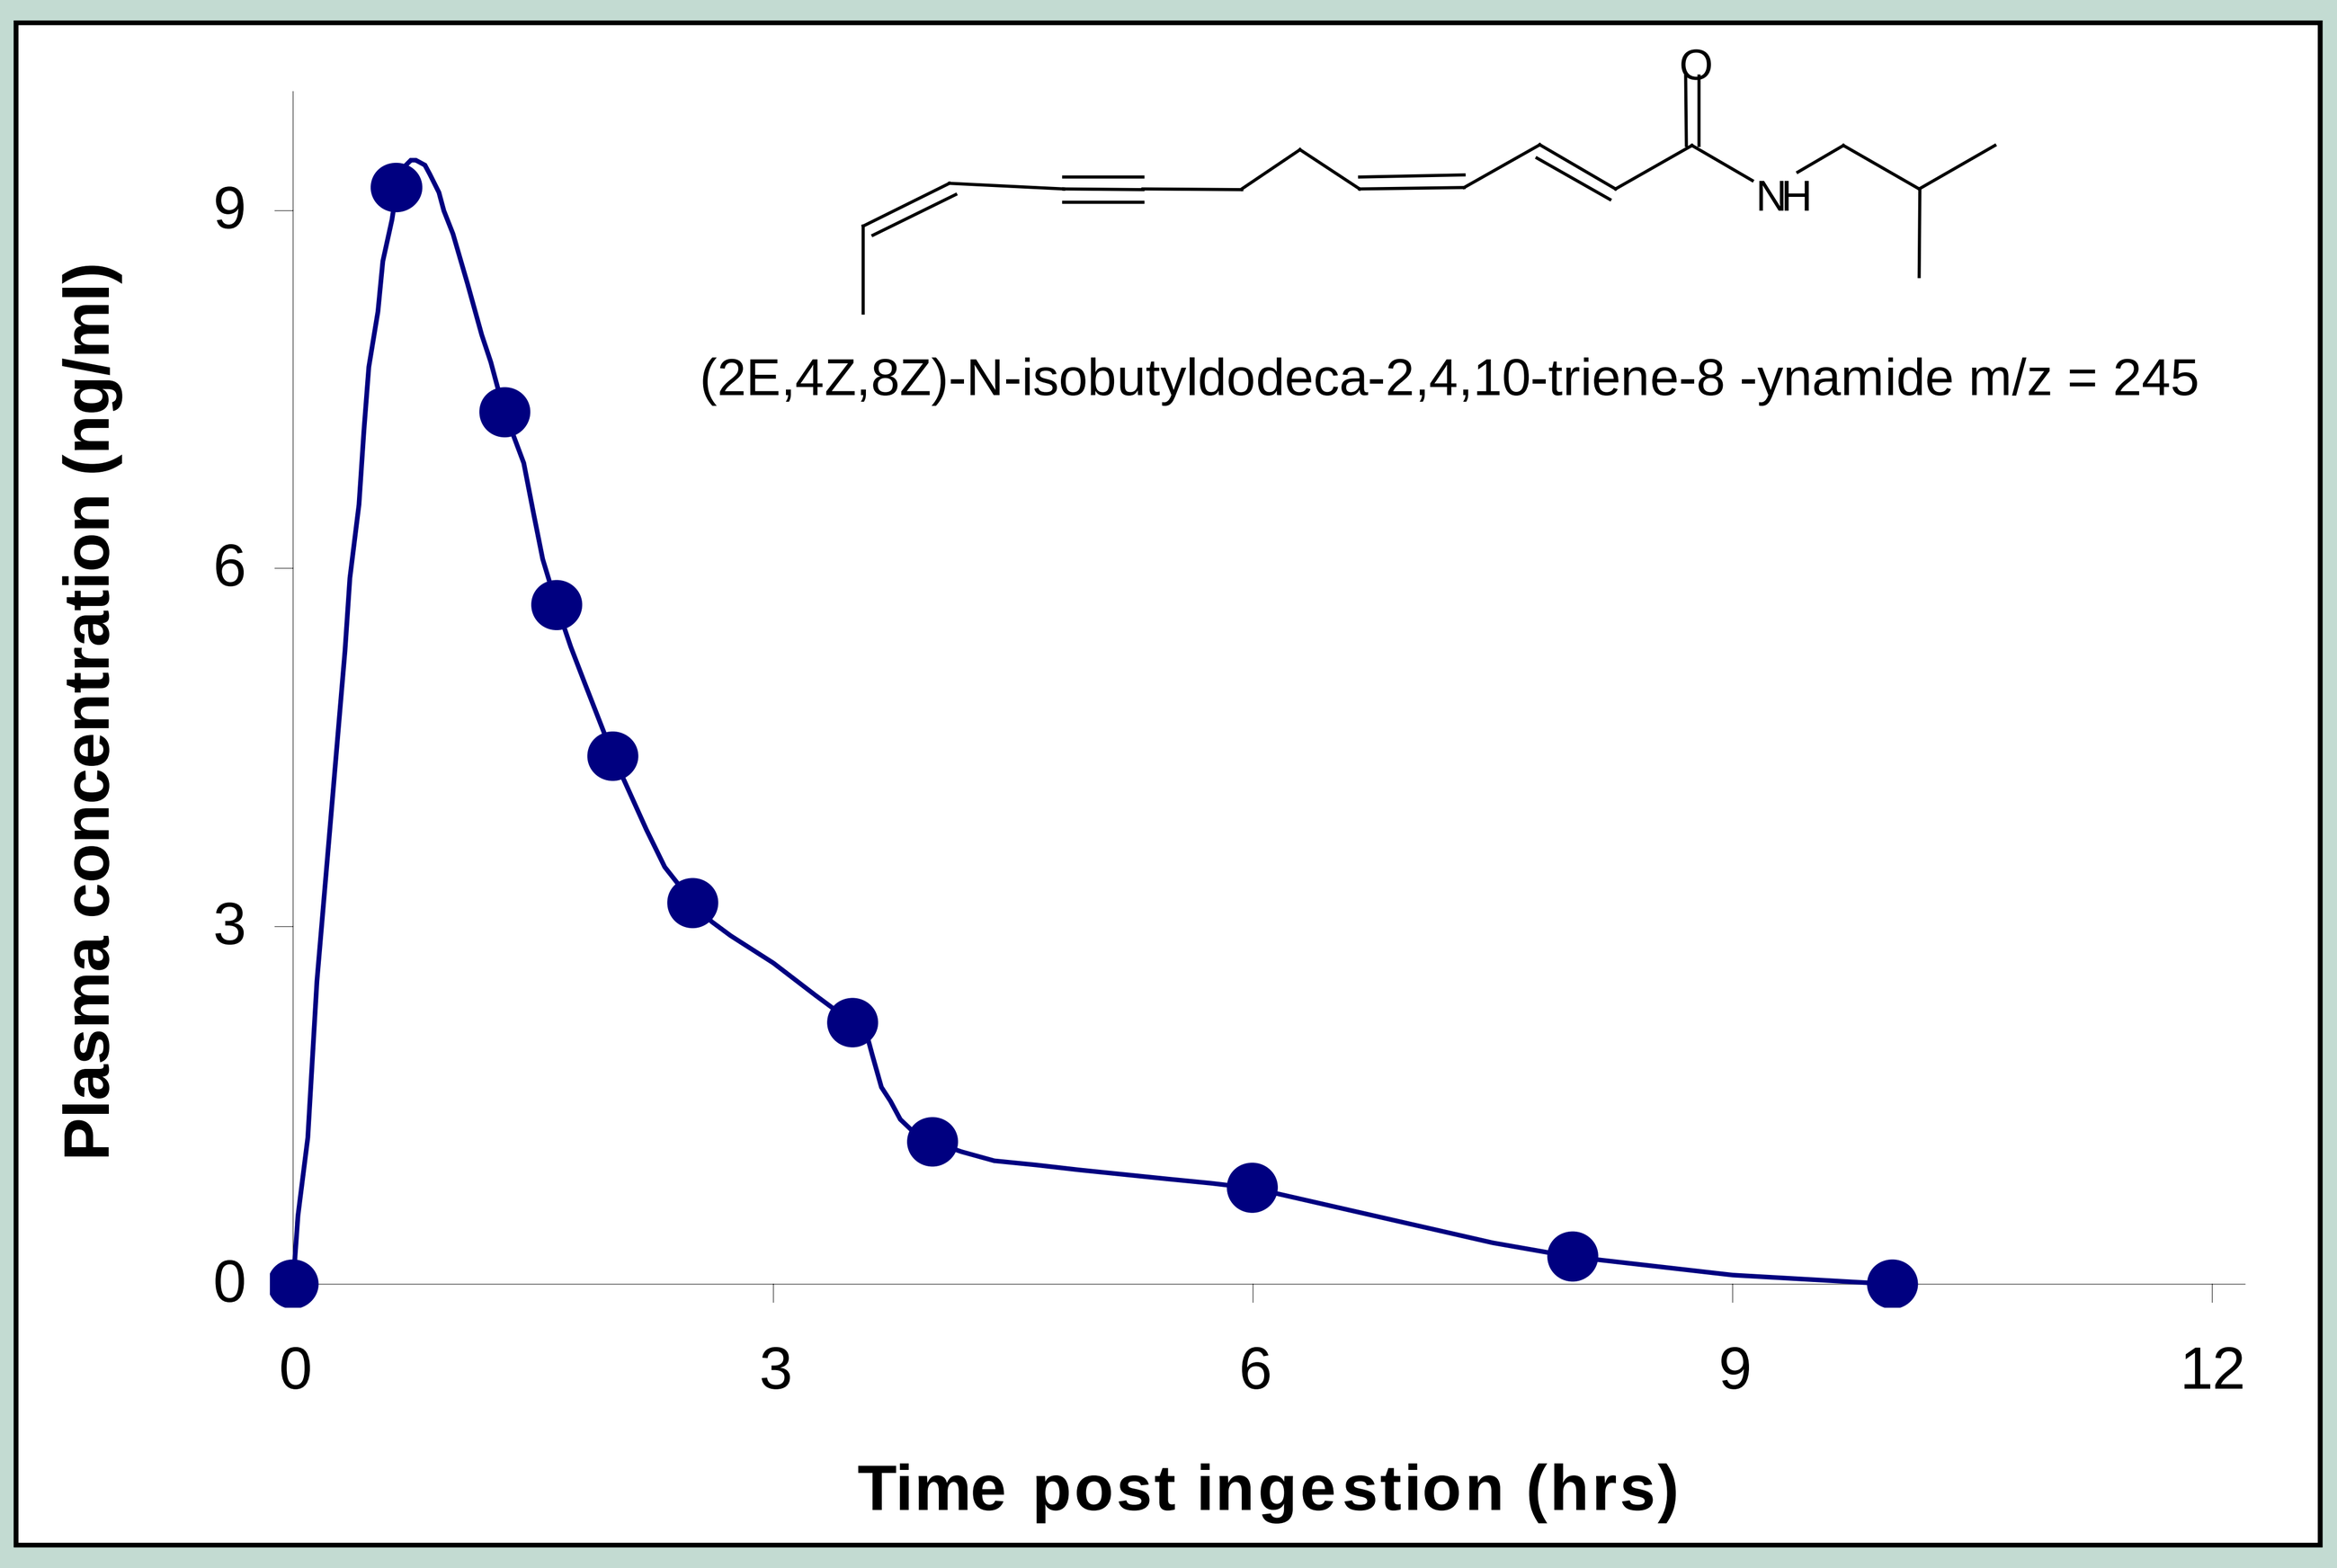
(2E,4Z,8Z)-N-isobutyldodeca-2,4,10-triene-8 -ynamide m/z = 245
Plasma concentration (ng/ml)
0
3
6
9
0
3
6
9
12
Time post ingestion (hrs)

Figure 2: Plasma alkylamide (m/z = 245) concentration.

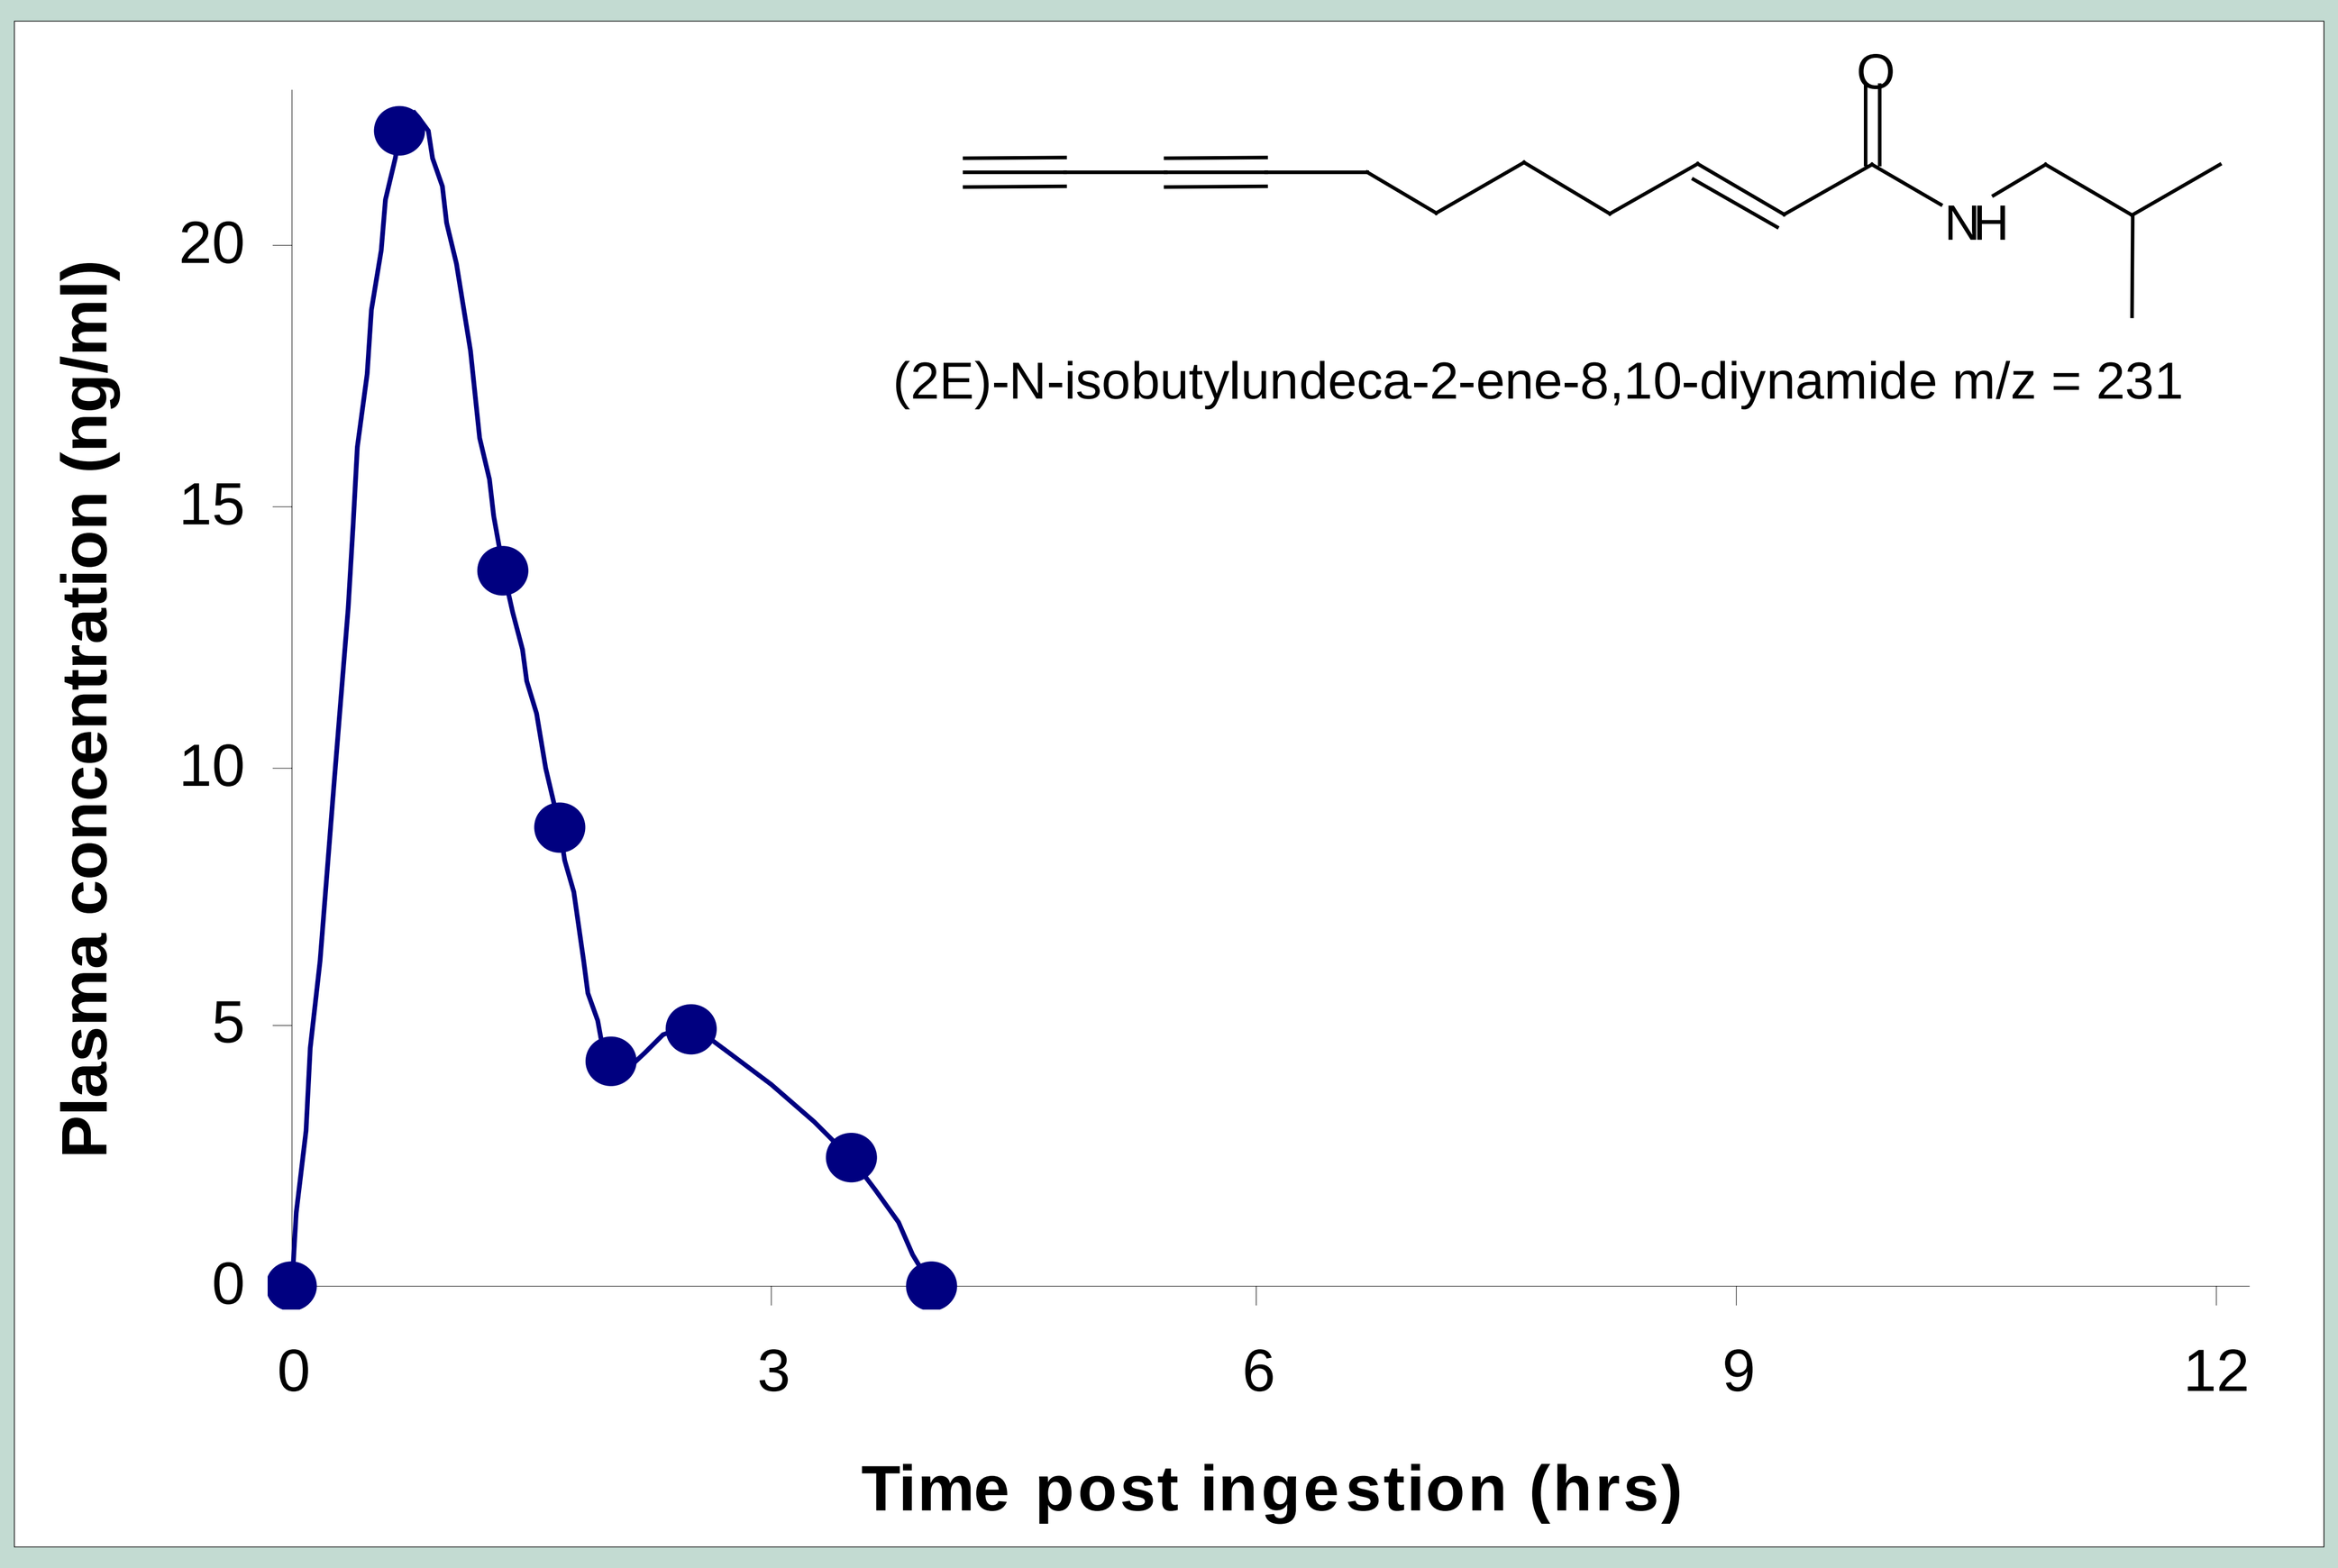
(2E)-N-isobutylundeca-2-ene-8,10-diynamide m/z = 231
Plasma concentration (ng/ml)
0
5
10
15
20
0
3
6
9
12
Time post ingestion (hrs)

Figure 3: Plasma alkylamide (m/z = 231) concentration.

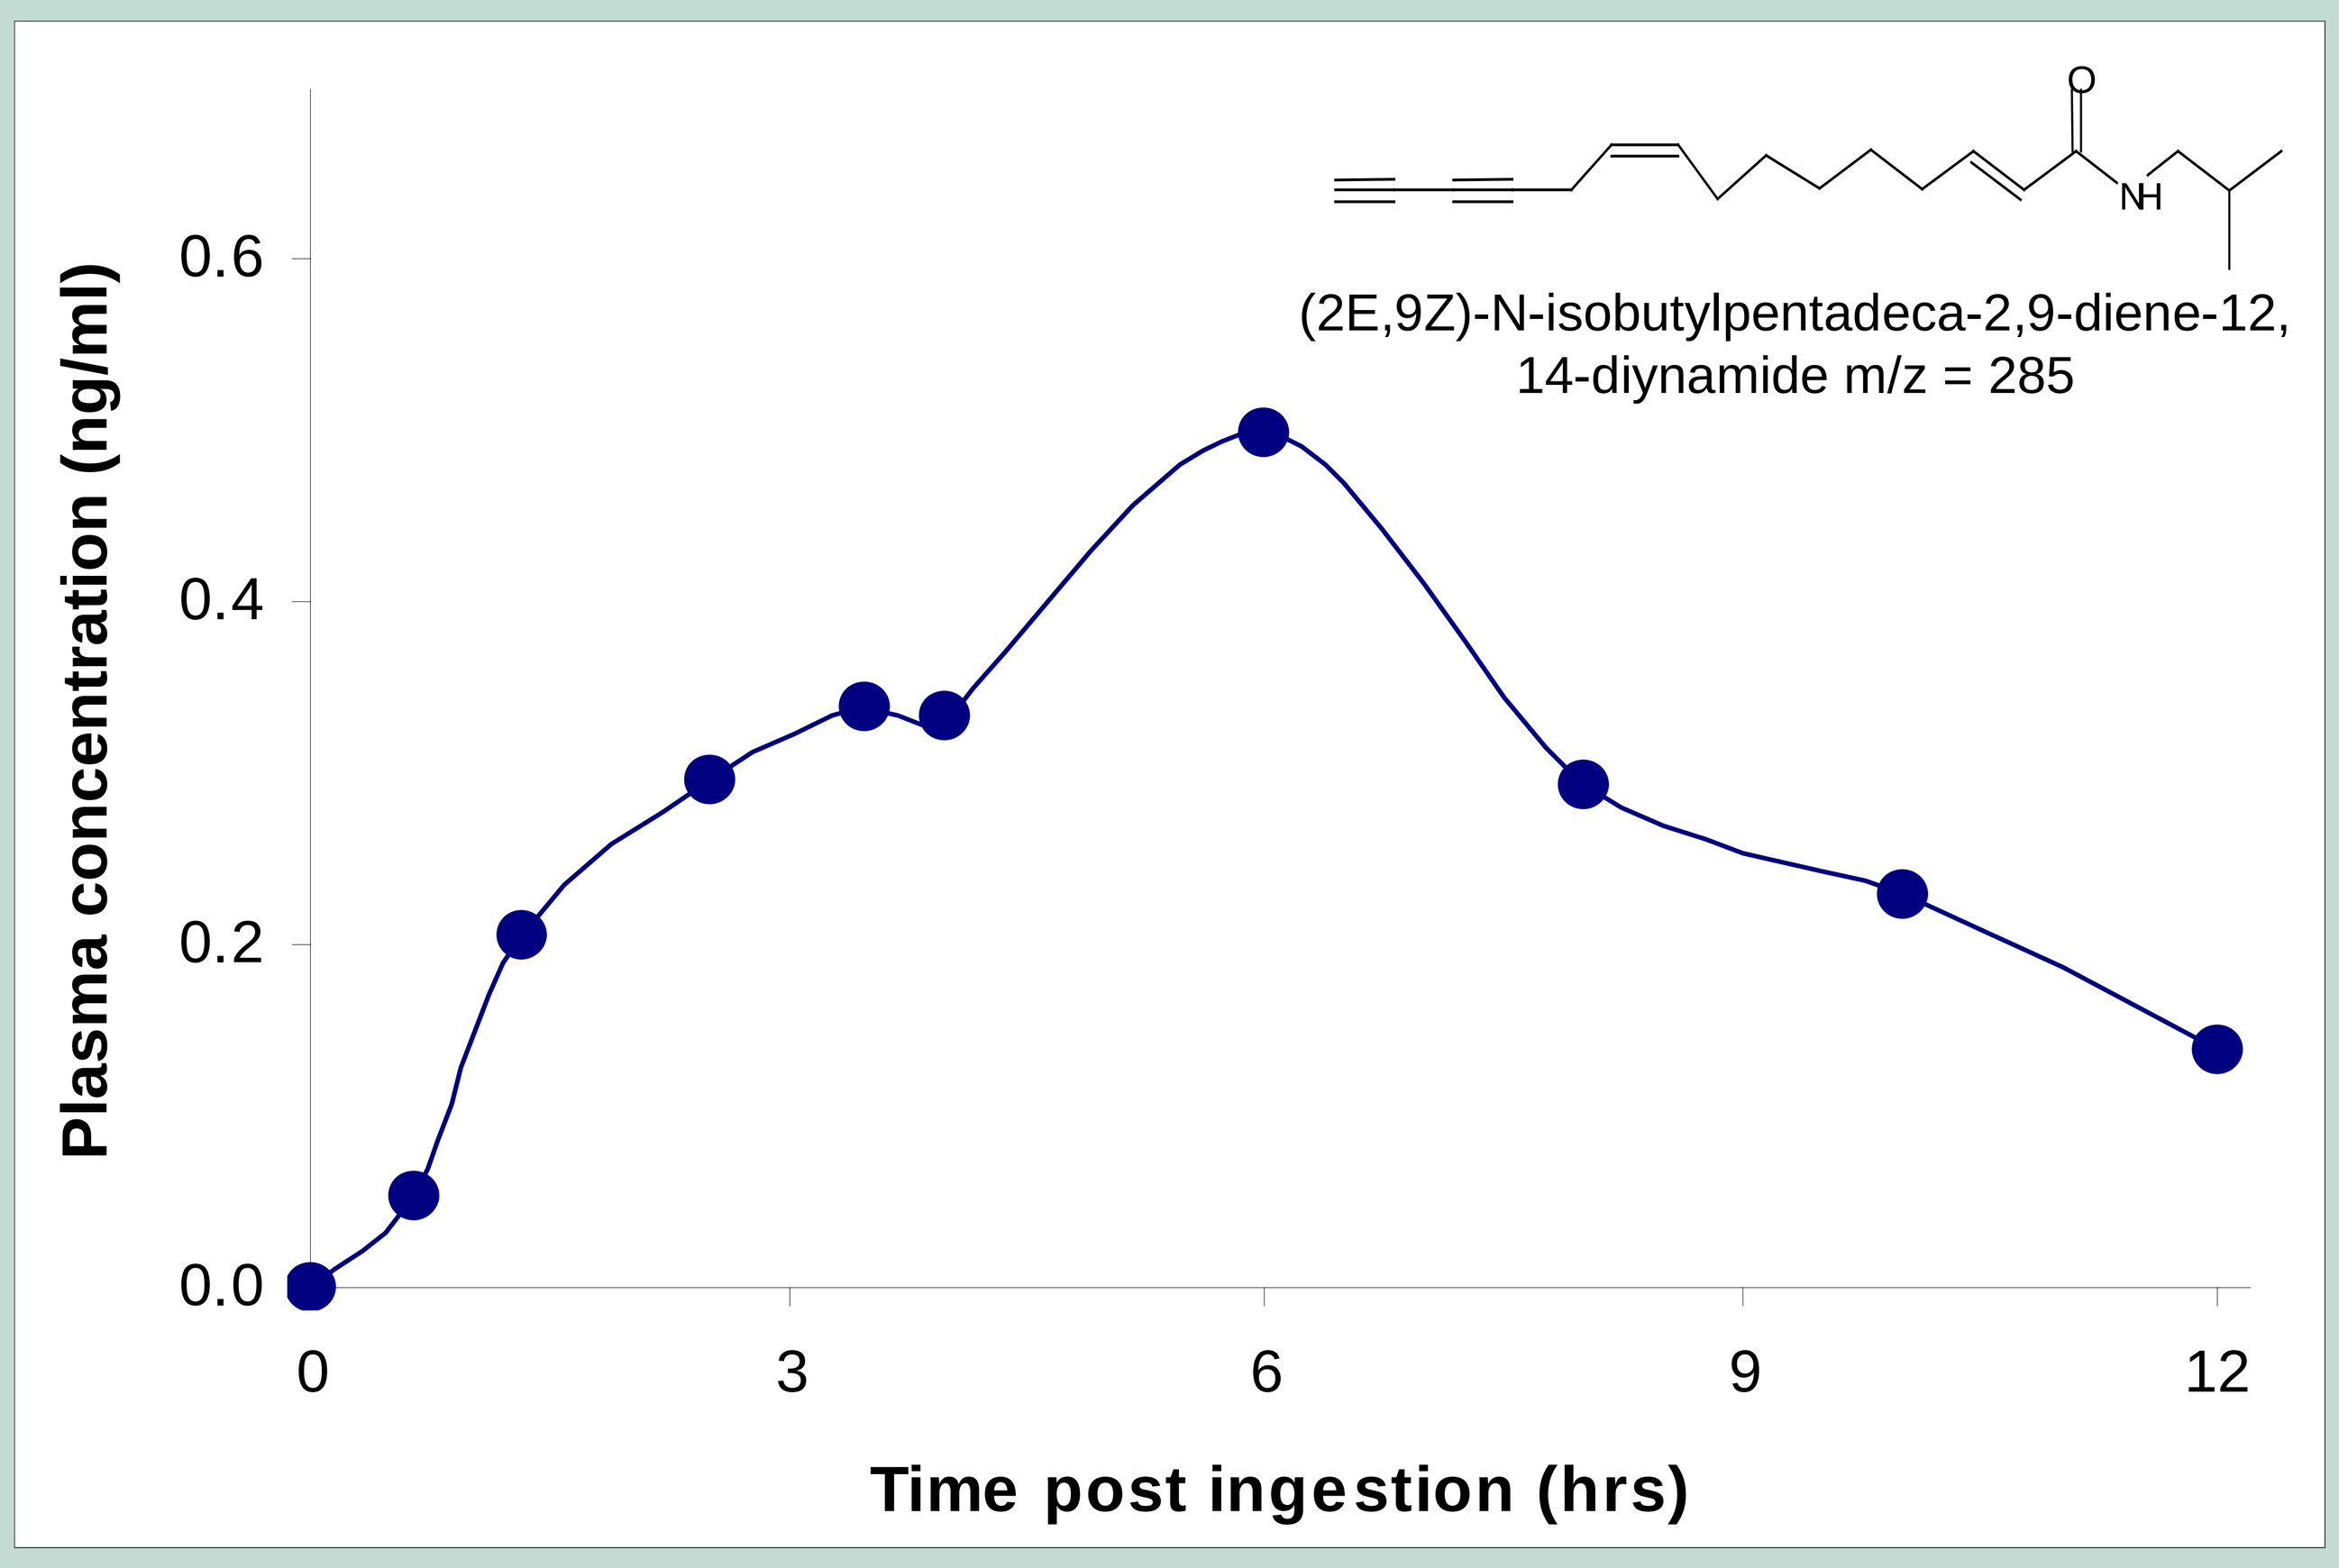
(2E,9Z)-N-isobutylpentadeca-2,9-diene-12,
14-diynamide m/z = 285
Plasma concentration (ng/ml)
0.0
0.2
0.4
0.6
0
3
6
9
12
Time post ingestion (hrs)

Figure 4: Plasma alkylamide (m/z = 285) concentration.

Methods

Healthy fasted volunteers ingested 4 tablets and serial blood samples were then taken over the next 12 hours. Blood samples were centrifuged and the plasma separated. Plasma was kept frozen at -20°C until analysed for the presence of alkylamides and caffeic acid conjugates.

Briefly, plasma was mixed volume for volume with ammonium acetate containing internal standards. This was then passed through an SPE cartridge for separation of either the alkylamides or caffeic acid conjugates. Eluates were then dried under air at 37°C and reconstituted in a small volume of ethanol. Alkylamides were quantitated using LC-MS and caffeic acid conjugates using HPLC-PDA.

Results

Alkylamide	**247**	**245**	**231**	**285**	**CA**
C_{max} (ng/ml plasma)	82.5	9.20	22.2	0.50	ND
T_{max} (hours)	0.66	0.66	0.66	6.00	ND
$T_{1/2}$ (elimination)	3.36	1.52	2.95	3.38	ND
AUC (ng.hr/ml)	198	21.1	32.0	3.48	ND

Table 1: Pharmacokinetic parameters of Echinacea components after ingestion.

Data for alkylamides of molecular weights 247, 245, 231 and 285 and the caffeic acid conjugate cichoric acid (CA) are shown. Data is the average of two individuals.

C_{max} = maximum plasma concentration; T_{max} = time at which C_{max} occurs; $T_{1/2}$ (elimination) = the time taken for the plasma concentration of a given compound to decrease by one half in the elimination phase of the pharmacokinetic curve; AUC = area under the curve - an indication of the total uptake for each compound; ND = not determined as no compound could be detected.

- **Alkylamides were detected in plasma 20 minutes after tablet ingestion.**
- **Most of the alkylamides found in Echinacea were detected in plasma.**
- **Alkylamides remained detectable in plasma for up to 12 hours.**
- **Caffeic acid conjugates are not bioavailable.**

Summary

Alkylamides and not caffeic acid conjugates are found in human plasma after ingestion of Echinacea tablets.

These data are consistent with the dosing regimen for MediHerb Echinacea Premium Tablets already recommended: One tablet three times daily.

Acknowledgments: This work was funded by AUSINDUSTRY through a Biotechnology Innovation Fund Grant (No. BIF02651) and MediHerb Pty. Ltd.

References:
[1] Dietz B, Heilmann J and Bauer R. (2001) Absorption of dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamides after oral application of *Echinacea purpurea* tincture. Planta Med. (2001) 67: 863-864

[2] Matthias A, Blanchfield JT, Penman KG, Toth I, Lang C-S, De Voss JJ and Lehmann RP. Permeability studies of alkylamides and caffeic acid conjugates from Echinacea using a Caco-2 cell monolayer model. J. Clin. Pharm. Therapeut. (2004) 29: 7-13

MEDIHERB®